In vitro immunomodulatory comparison of Maternal vs Fetal Placenta-derived cells

Andrea Papait[*1], Marta Magatti*[*1], Elsa Vertua[1], Antonietta Silini[1], Racheli Ofir[2], Ornella Parolini [1,3]*
**co-first authors*
[1] Centro di Ricerca E. Menni, Fondazione Poliambulanza, Brescia, Italy, [2] Pluristem LTD, Haifa, Israel, [3] Istituto di Anatomia Umana e Biologia Cellulare, Università Cattolica del Sacro Cuore, Rome, Italy

Background

In the last decade, placenta-derived mesenchymal stromal cells have acquired great attention for their immunomodulatory properties and poor immunogenicity making them suitable for allogeneic transplantation. Although mesenchymal stromal cells isolated from different areas of the placenta show similar characteristics, they also have peculiar properties that make them distinct. Here we compared cells derived from full term placenta distinguishing them on the basis of their maternal or fetal origin, and we evaluated their immunomodulatory properties. Experiments were performed with two batches of maternal-derived cells (PLacenta eXpanded mesenchymal like adherent stromal cells (PLX) R08 an R12), and two types of fetal-derived cells (PLX-R18 and human amniotic mesenchymal stromal cells (hAMSC). PLX cells were developed by Pluristem Therapeutics, hAMSC were isolated in our laboratory [1.2].

1. Fetal and maternal cells share similarities with MSC profile

	Markers	R08	R12	R18	hAMSC
hMSC	CD90	86,4	77,85	93,39	85,9
	CD105	60	85,43	54,8	66,7
	CD73	90,3	77,22	17,18	61,6
	CD13	70,89	61,65	72,91	68,5
Epithelial	CD324	0,34	0,12	0,17	0,2
	CD326	0,52	0,81	0,1	0,5
Haematopoietic	CD45	8,64	1,05	0,68	3,5
	CD66b	0	0	0,03	0,0
	CD34	0,49	0,57	0,15	0,4
Pericytes	CD107a	29,82	1,19	0,69	10,6
	CD146	29,99	6,84	8,22	15,0
	CD140b	92,13	40,77	84,61	72,5
Histocompatibility	HLA-ABC	25,59	9,42	15,27	16,8
	HLA-DR	0,09	0,03	0,03	0,1

Table 1. Informative surface markers expressed by PLX cells. *Phenotypic analysis of MSCs derived from different batches of PLX-PAD (maternal derived cells) vs fetal derived cells (R18/hAMSC). Results are expressed as % of positive cells. We compared different batches of maternal derived cells (PLX-PAD R08 and R12) cells with the expression profile of fetal derived cells (PLX-R18, hAMSC isolated in house).*
Table 1 summarizes the data from the CD screen. PLX-PAD as well as PLX-R18 cells showed the consensus expression profile of MSC, such as CD 73$^+$ 90$^+$ 105$^+$ CD45$^-$34$^-$.

2. Effect of Placental cells on T Lymphocyte Proliferation

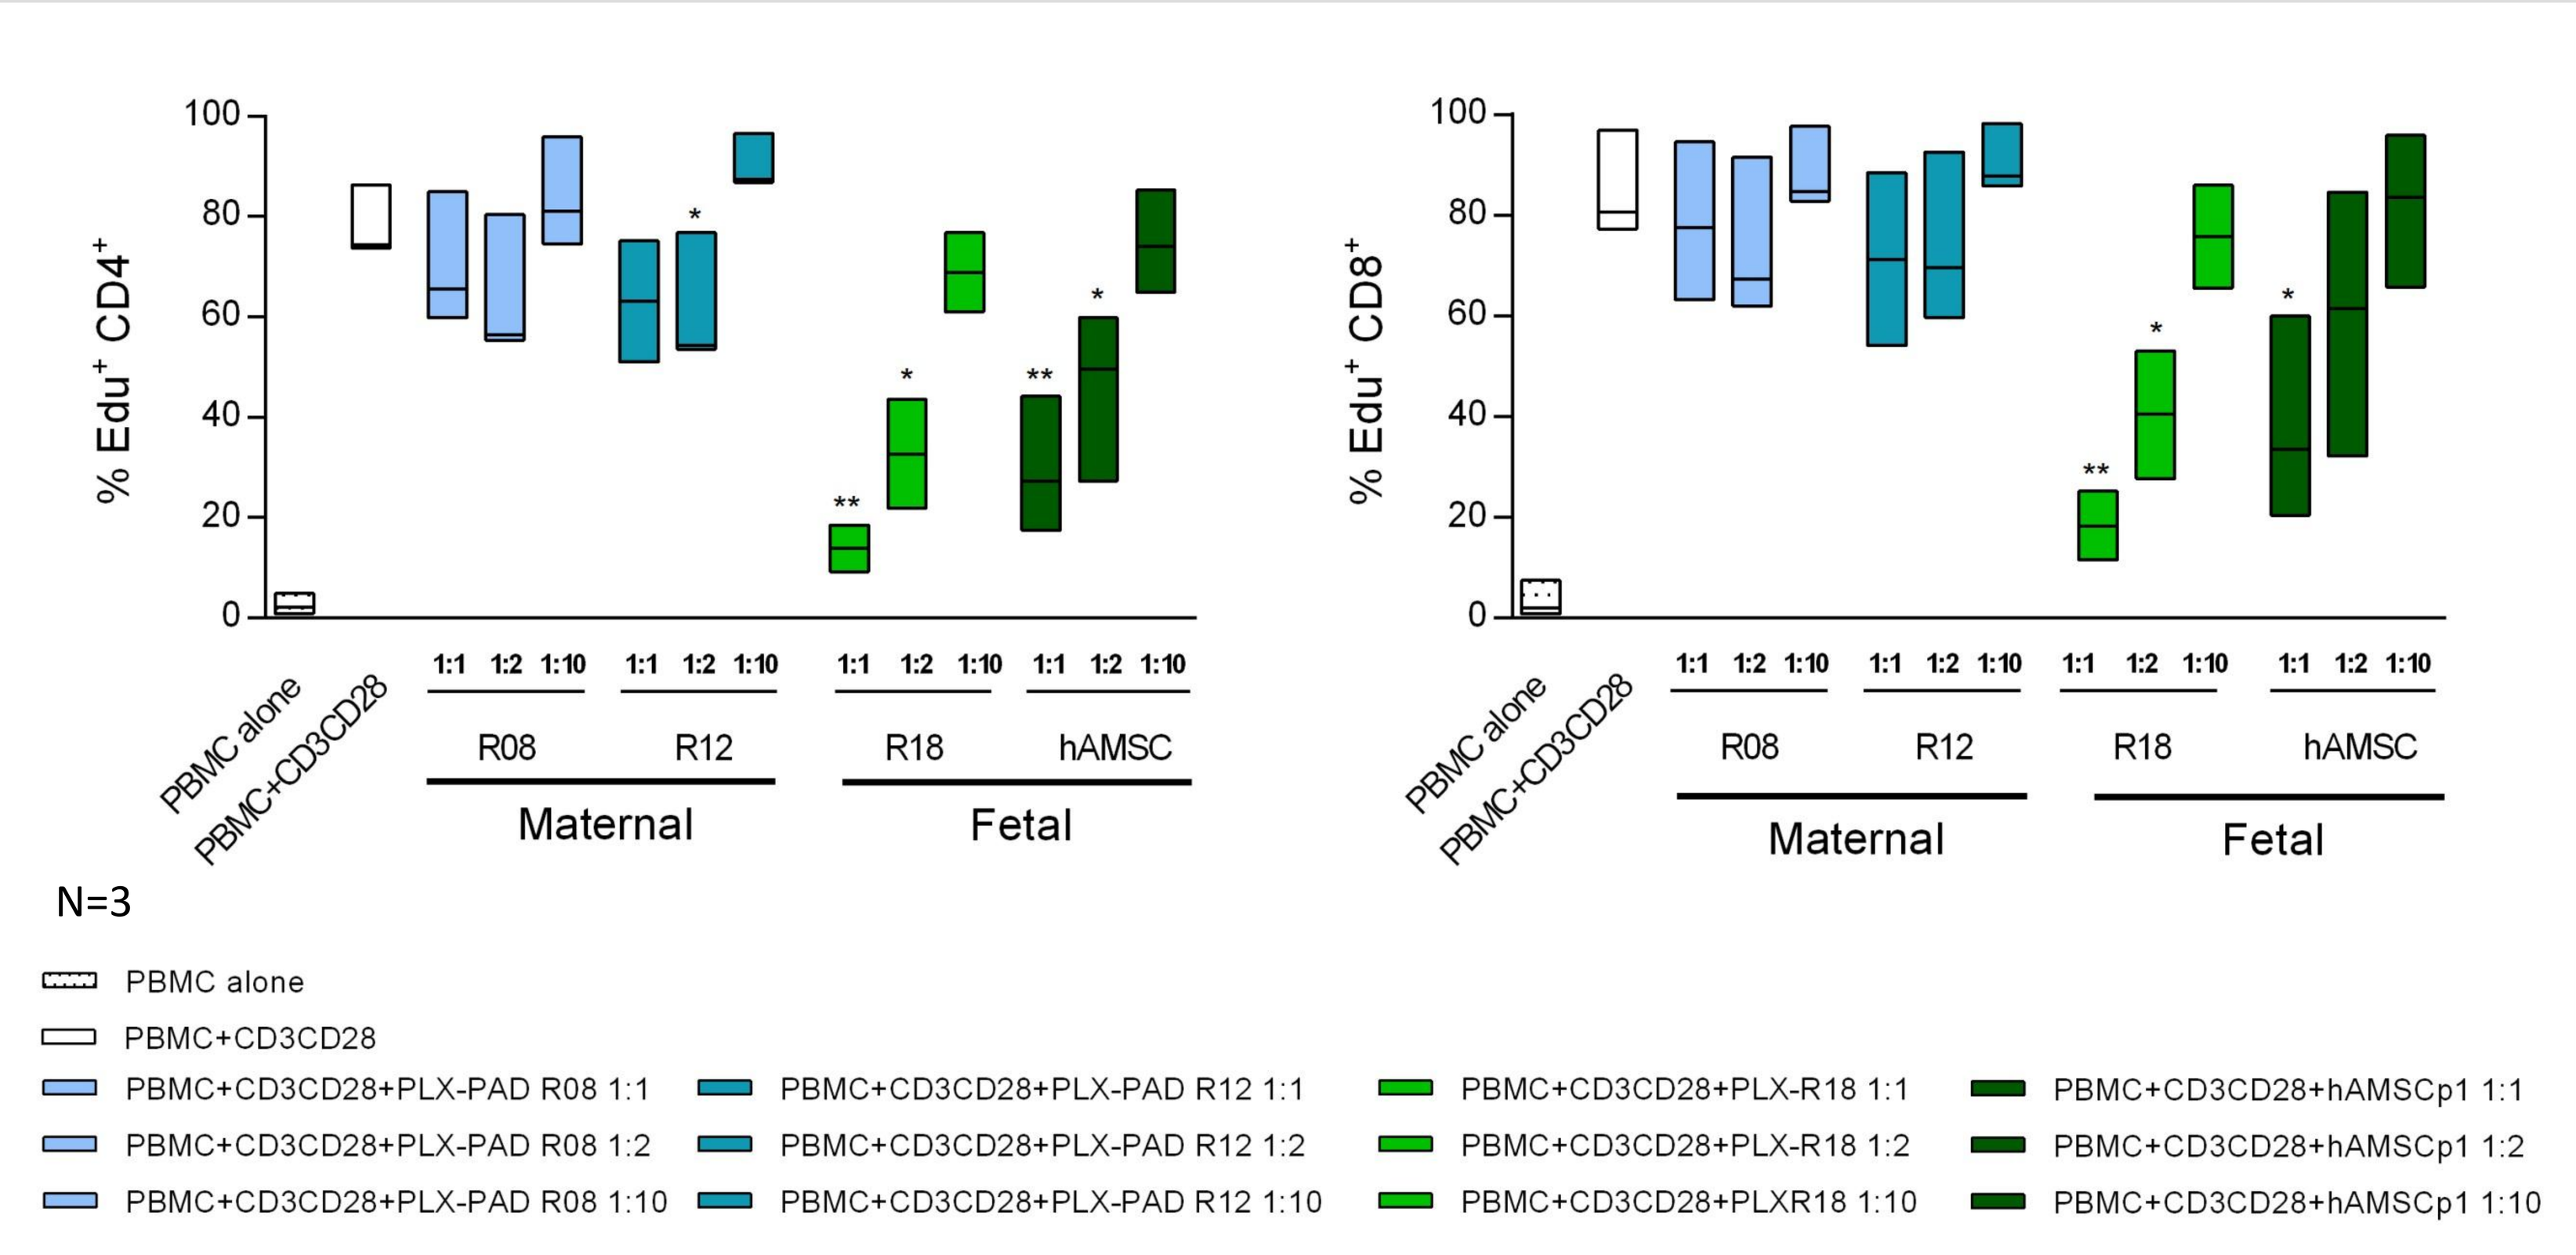
% Edu+ CD4+
% Edu+ CD8+
PBMC alone
PBMC+CD3CD28
R08
R12
R18
hAMSC
Maternal
Fetal
N=3
PBMC+CD3CD28+PLX-PAD R08 1:1
PBMC+CD3CD28+PLX-PAD R08 1:2
PBMC+CD3CD28+PLX-PAD R08 1:10
PBMC+CD3CD28+PLX-PAD R12 1:1
PBMC+CD3CD28+PLX-PAD R12 1:2
PBMC+CD3CD28+PLX-PAD R12 1:10
PBMC+CD3CD28+PLX-R18 1:1
PBMC+CD3CD28+PLX-R18 1:2
PBMC+CD3CD28+PLXR18 1:10
PBMC+CD3CD28+hAMSCp1 1:1
PBMC+CD3CD28+hAMSCp1 1:2
PBMC+CD3CD28+hAMSCp1 1:10

Figure 2. Effect of placental cells on T lymphocyte proliferation. *Allogeneic PBMC (1 × 105) were stimulated with anti-CD3/CD28 antibodies (white box, second lane) in absence or in the presence of decreasing ratios of maternal (PLX-PAD R08 or R12) or fetal (PLX-R18 or hAMSC) derived cells. Cells were cultured for 3 days, and proliferation was assessed by Edu (ethynyldeoxyuridine) incorporation added during the final 18 h of culture. Results are expressed as percentage of proliferation, with the positive control represented by PBMC stimulated with CD3CD28 mAbs. Percentage of proliferation was calculated for both CD4+ and CD8+ T-lymphocyte cell subset. PBMC alone is representative of the basal level of proliferation in absence of stimulus and in the absence of MSC. Results show median+/- sem (∗, $p < 0.05$), N=3*

3. Effect of Placental cells on Lymphocyte Function (Cytotoxicity)

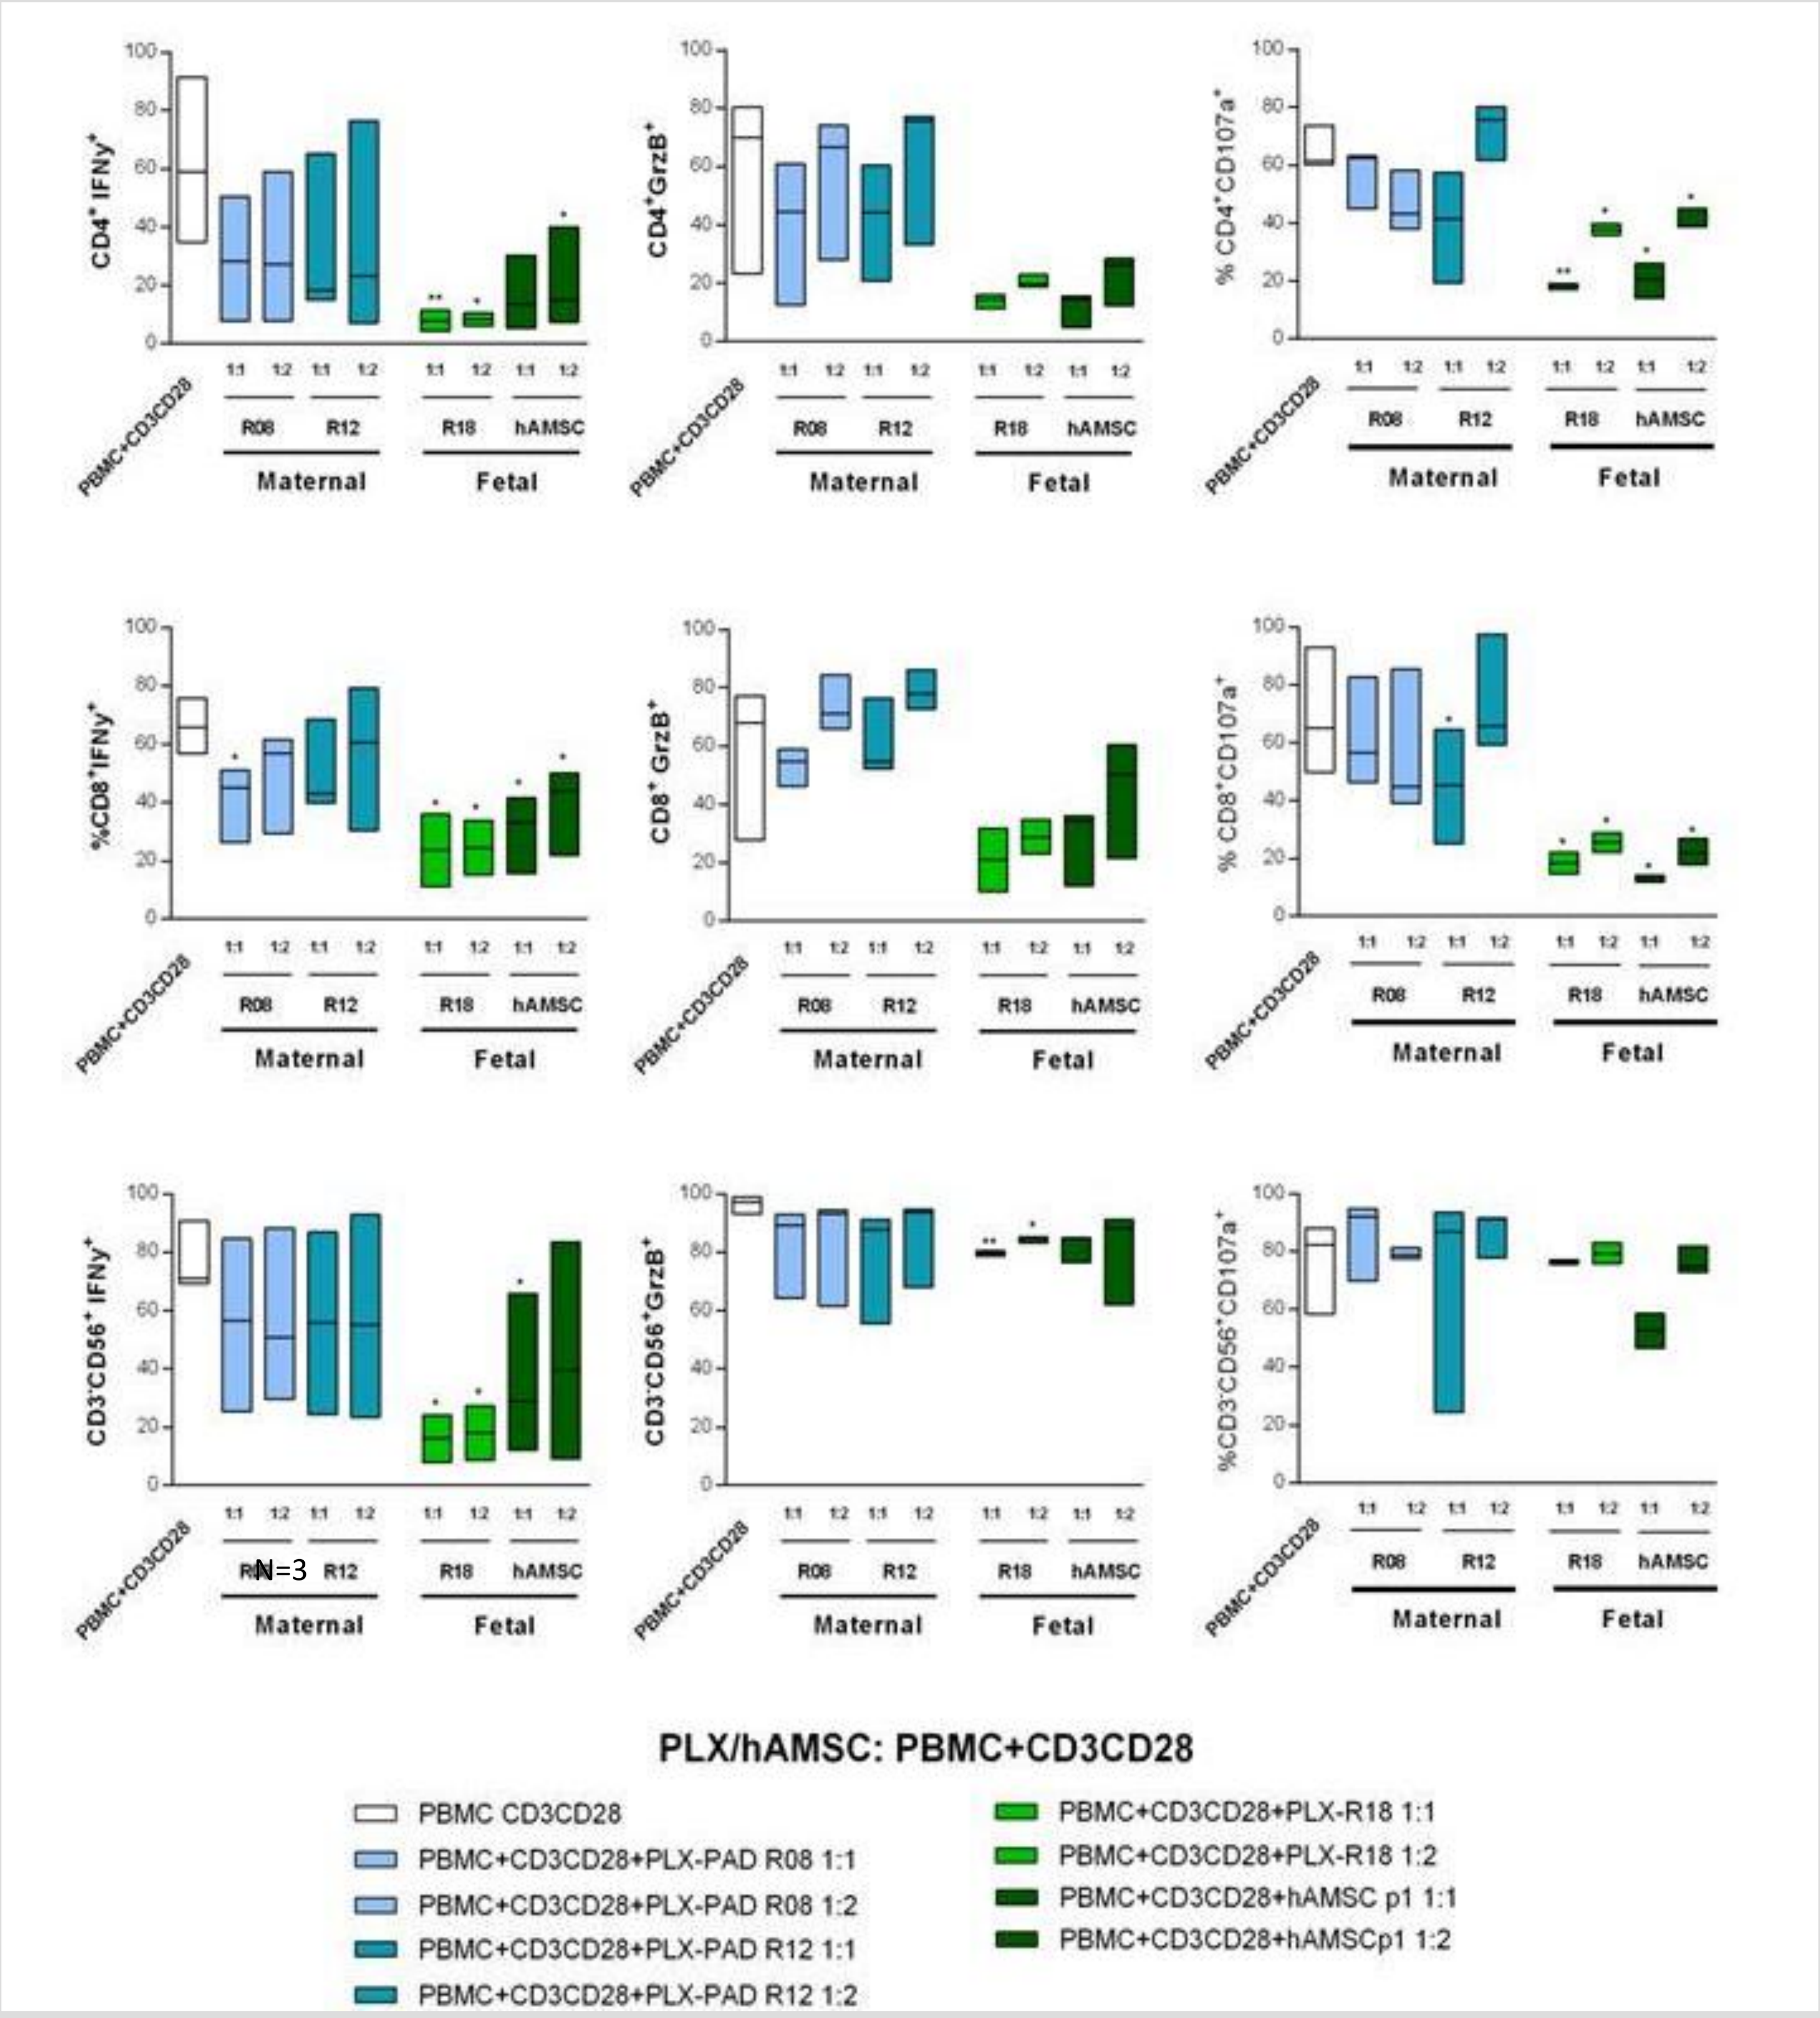
CD4+ IFNγ+
CD4+GrzB+
% CD4+CD107a+
%CD8+IFNγ+
CD8+ GrzB+
% CD8+CD107a+
CD3-CD56+ IFNγ+
CD3-CD56+GrzB+
%CD3-CD56+CD107a+
PBMC+CD3CD28
R08
R12
R18
hAMSC
Maternal
Fetal
N=3
PLX/hAMSC: PBMC+CD3CD28
PBMC CD3CD28
PBMC+CD3CD28+PLX-PAD R08 1:1
PBMC+CD3CD28+PLX-PAD R08 1:2
PBMC+CD3CD28+PLX-PAD R12 1:1
PBMC+CD3CD28+PLX-PAD R12 1:2
PBMC+CD3CD28+PLX-R18 1:1
PBMC+CD3CD28+PLX-R18 1:2
PBMC+CD3CD28+hAMSC p1 1:1
PBMC+CD3CD28+hAMSCp1 1:2

Figure 3. Cytotoxicity markers production by CD4+,CD8+ and NK cells-on interaction with MSCs. *IFNy. CD107a and Granzyme B production by CD3CD28 activated PBMC after interaction with MSCs. Allogeneic PBMC were incubated alone or in the presence of MSCs (maternal in grey and fetal derived in green) at 2 ratio (1:1 and 1:2). After 6 hours of culture in the presence of PMA+Ionomycin and monensin-containing GolgiStop, cells were collected, and staining was performed. To detect the frequency of IFN-y, CD107a and Granzyme B positive cells, CD4, CD8 and NK cells were stained with anti–CD56, anti-CD3, antiCD8, anti–IFN-y, antiCD107a and anti-GranzymeB mAbs and were analyzed by flow cytometry. Results show median+/- sem (∗, $p < 0.05$), N=3*

4. Effect of Placental cells on Monocyte differentiation towards APCs

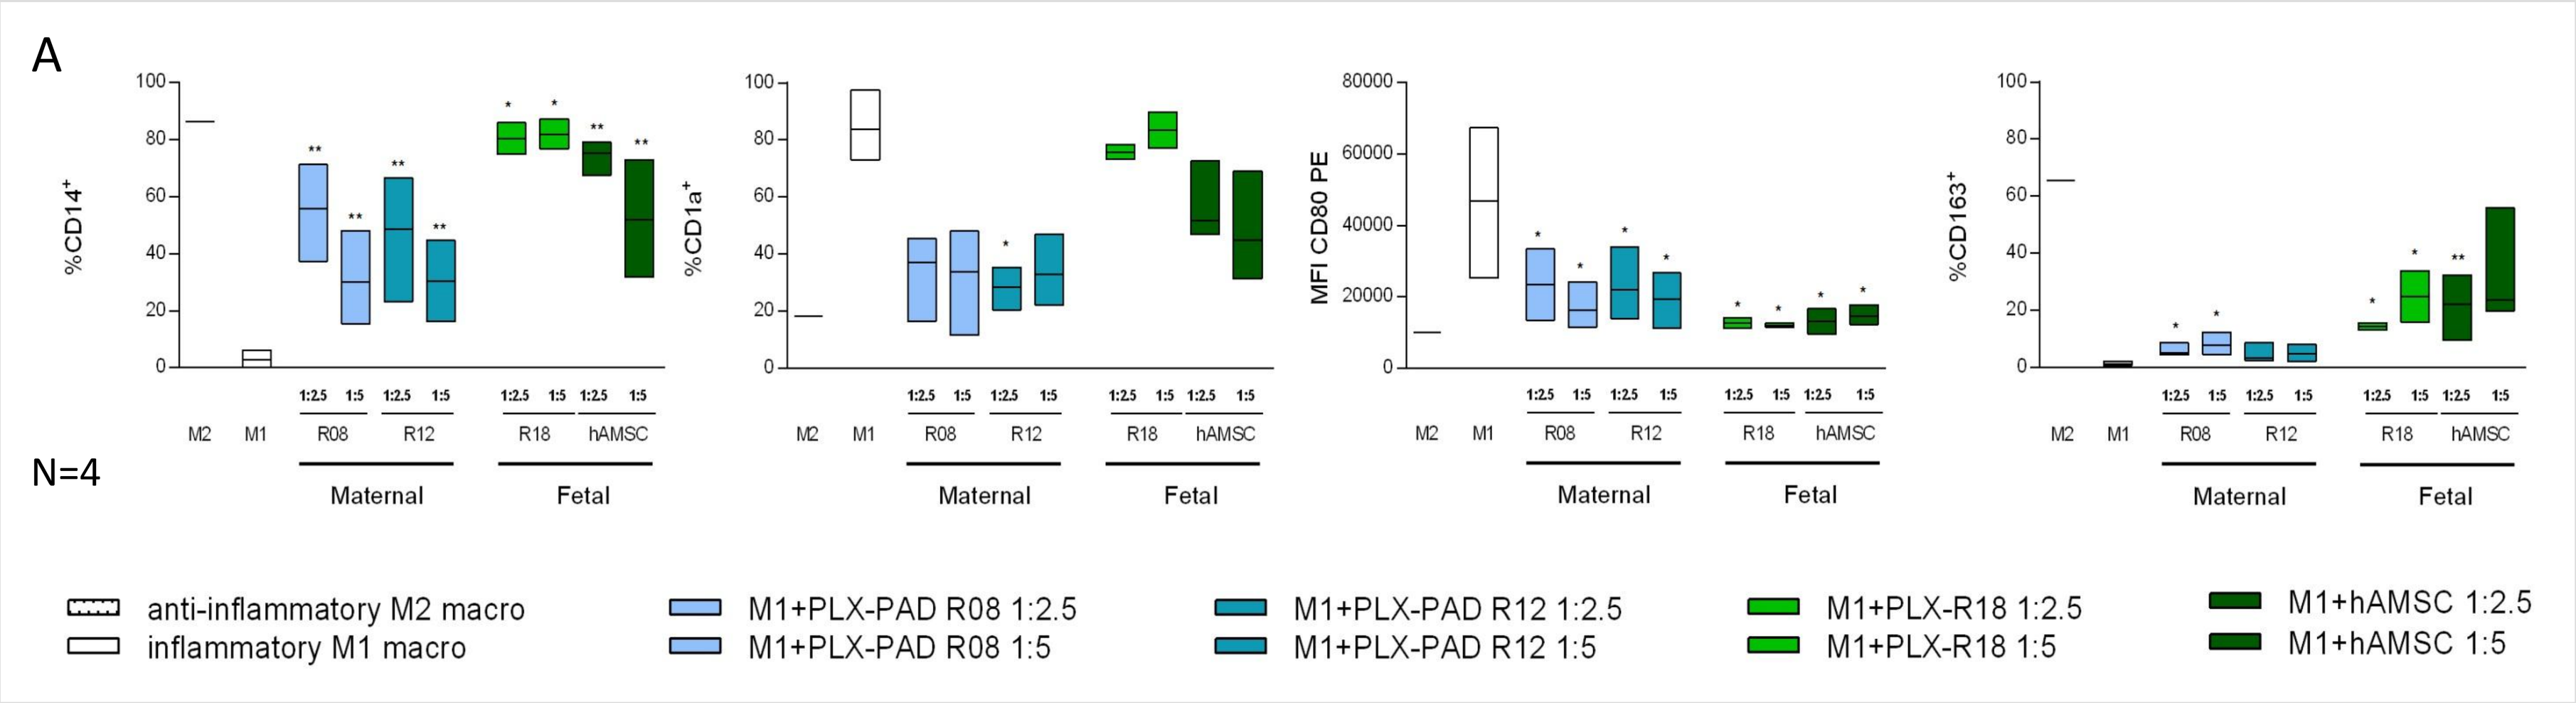
A
%CD14+
%CD1a+
MFI CD80 PE
%CD163+
M2
M1
R08
R12
R18
hAMSC
Maternal
Fetal
N=4
anti-inflammatory M2 macro
inflammatory M1 macro
M1+PLX-PAD R08 1:2.5
M1+PLX-PAD R08 1:5
M1+PLX-PAD R12 1:2.5
M1+PLX-PAD R12 1:5
M1+PLX-R18 1:2.5
M1+PLX-R18 1:5
M1+hAMSC 1:2.5
M1+hAMSC 1:5

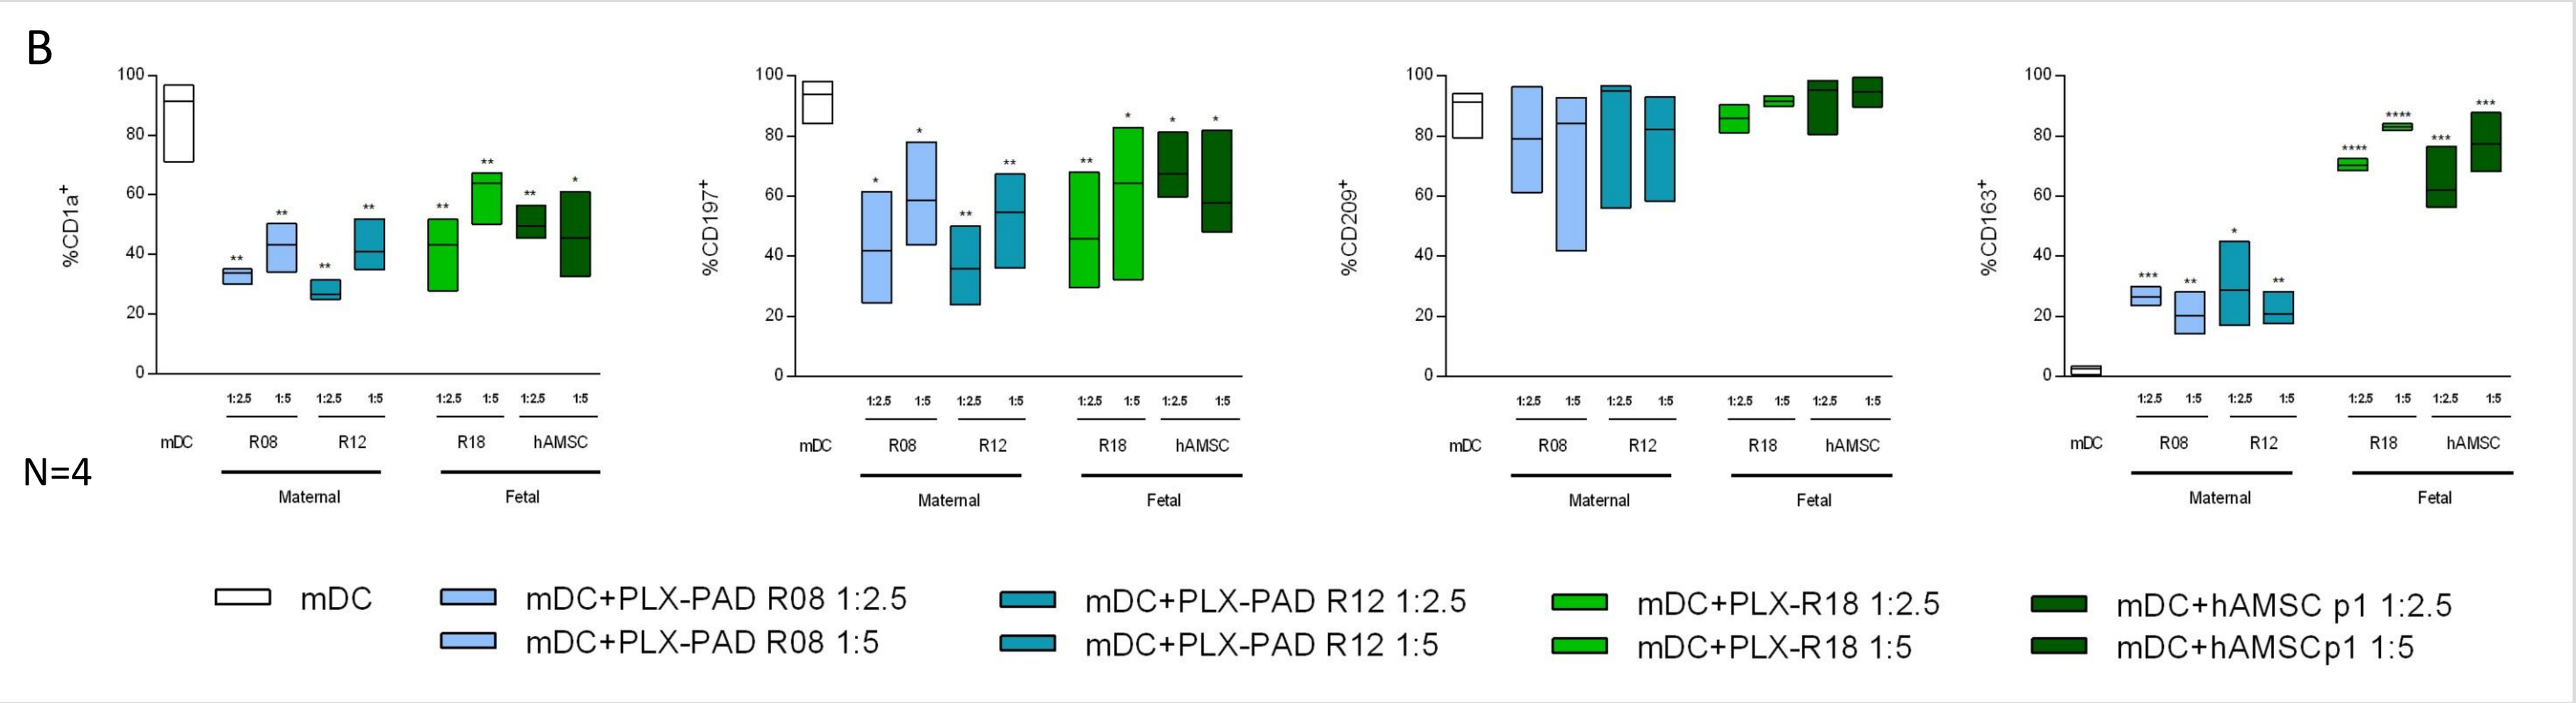
B
%CD1a+
%CD197+
%CD209+
%CD163+
mDC
R08
R12
R18
hAMSC
Maternal
Fetal
N=4
mDC
mDC+PLX-PAD R08 1:2.5
mDC+PLX-PAD R08 1:5
mDC+PLX-PAD R12 1:2.5
mDC+PLX-PAD R12 1:5
mDC+PLX-R18 1:2.5
mDC+PLX-R18 1:5
mDC+hAMSC p1 1:2.5
mDC+hAMSCp1 1:5

Figure 4. Effect of placental cells on differentiation of monocytes to APC *Phenotypic analysis of isolated CD14+ monocytes that were induced to differentiate into M1 macrophages (A) or mature dendritic cells (mDC) (B) in the absence or presence of maternal (PLX-PAD R08 or R12) or fetal derived (PLX-R18 or hAMSC) from the beginning of the culture period. Monocyte differentiation towards M1 macrophages was performed with GM-CSF fro 4 days followed by the administration of INF-γ+LPS for other 2 days (A). mDC differentiation was assessed with GM-CSF+IL-4 for 4 days followed by+ LPS for 2 days. M2 macrophages used as internal control to assess the level of expression of M2 markers as CD163 and CD14 were differentiated with M-CSF for 4 days followed by 2 days with + IL-4+LPS (A, first lane). After culture, expression of CD1a, CD14, CD197, CD80, CD209 and CD163 was evaluated by flow cytometry . Results show median+/- sem (∗, $p < 0.05$), N=4*

Conclusions

- **PLX cells have the phenotypical profile of hMSC**

For the immunomodulatory properties

- **Both PLX cells (PLX-PAD and PLX-R18), at ratio 1:1 reduce T cell proliferation, but fetal derived cells are more prone to inhibit T lymphocyte proliferation upon stimulation**
- **Both PLX cells (PLX-PAD and PLX-R18), at ratio 1:1 reduce the expression of cytotoxicity markers , but fetal-derived cells (PLX-R18 and hAMSC) were more prone to impact on the cytotoxicity**
- **Both PLX cells (PLX-PAD and PLX-R18), at ratio 1:1 interfere with monocyte differentiation towards M1 macrophages and mDC**

Our findings show that MSCs and MSC like cells from both fetal and maternal sources have similar basic biological properties, but fetal-derived cells (PLX-R18 and hAMSC) present enhanced immunomodulatory effects suggesting the relevance of the source used

References:

1. Magatti, M., et al., *Human amnion mesenchyme harbors cells with allogeneic T-cell suppression and stimulation capabilities.* Stem Cells, 2008. **26**(1): p. 182-92.1.
2. Parolini, O., et al., *Concise review: isolation and characterization of cells from human term placenta: outcome of the first international Workshop on Placenta Derived Stem Cells.* Stem Cells, 2008. **26**(2): p. 300-11.

Acknowledgement:

The authors would like to thank the physicians and midwives of the Department of Obstetrics and gynecology of Fondazione Poliambulanza-Istituto Ospedaliero, Brescia (Italy), the Regenerative Medicine Research Center (CROME) of Università Cattolica del Sacro Cuore, and all the mothers who donate their baby's placenta to make this research possible. This work contributes to the COST Action CA17116 International Network work for Translating Research on Perinatal Derivatives into Therapeutic Approaches (SPRINT), supported by COST (European Cooperation in Science and Technology). The work presented in this poster was supported by Fondazione Poliambulanza and the European Union's Horizon 2020 research and innovation programme under grant agreement No 779293.